Study ALLN-177-206: Case Series of First Four Subjects with EH and Advanced CKD Treated with Reloxaliase

Felix Knauf[1], John C. Lieske[2], Anja C. Pfau[1], Danica Grujic[3], Kristine E. Bernard[3], Annamaria T. Kausz[3]

[1] University Hospital Charité Berlin, Berlin, Germany; [2] Mayo Clinic, Rochester, MN, USA; [3] Allena Pharmaceuticals, Inc., Newton, MA, USA

Background

- Enteric hyperoxaluria (EH), a serious metabolic disorder, results from fat malabsorption due to GI surgery and other GI conditions. Despite being a major risk factor for kidney stones it also can lead in severe cases to CKD and ESRD
 - Oxalate nephropathy secondary to EH:[1]
 - » Most patients remained dialysis-dependent or required kidney transplant, with 33% mortality rate
- No effective pharmacological treatment exists and dietary restriction, calcium supplementation, and high liquid intake is the only therapeutic approach. Difficult to sustain and/or suboptimal effect
- Reloxaliase is an oral enzyme, oxalate-specific therapy that degrades oxalate in the GI tract
- This basket[2] study is enrolling patients ≥12 years of age to examine the potential of reloxaliase to reduce both UOx and POx

Hypothesis

- Declining kidney function leads to oxalate accumulation in plasma (hyperoxalemia) and body (systemic oxalosis)
- With declining kidney function, the GI tract has the potential to play a more significant role in reduction of oxalate burden
- By degrading oxalate in the GI tract, reloxaliase may be able to reduce oxalate burden in the body measured by UOx and POx reduction

Open-Label 3-Month Basket Study

Study Objectives: Evaluate reloxaliase efficacy and tolerability in subjects with severe enteric hyperoxaluria (EH with CKD/ESRD and hyperoxalemia)

End Point: Change from baseline in plasma oxalate (POx) and urine oxalate (UOx/d)

Rationale:
- Basket study in severe forms of hyperoxaluria: EH with CKD, dialysis or post kidney transplant with hyperoxalemia, PH1-3
- Pilot-testing use of plasma oxalate (POx) in a multicenter trial for potential future studies
- First exposures in dialysis, PH, and adolescents

Screening and Baseline: Up to 4 Weeks	Treatment Period: 3 Months			Follow-Up: 4 Weeks
Key Entry Criteria	**Week 4**	**Week 8**	**Week 12**	
• EH and PH: ≥12 years • EH: UOx ≥ 40 mg/24h, POx > 5 µmol/L, and GFR < 45 mL/min/1.73m^2 n≈up to 20	Reloxaliase 7,500 u with meal/snacks 5xd for 12 weeks, up to 37,500 u/d			
2 x POx 3 x UOx/d	1 x POx 2 x UOx/d	1 x POx 2 x UOx/d	1 x POx 2 x UOx/d	1 x POx 2 x UOx/d

Self-selected diet recorded in diaries; blood samples collected in the morning after an overnight fast or prior to new dialysis sessions

Urine and Plasma Oxalate Reduction in Subjects with CKD

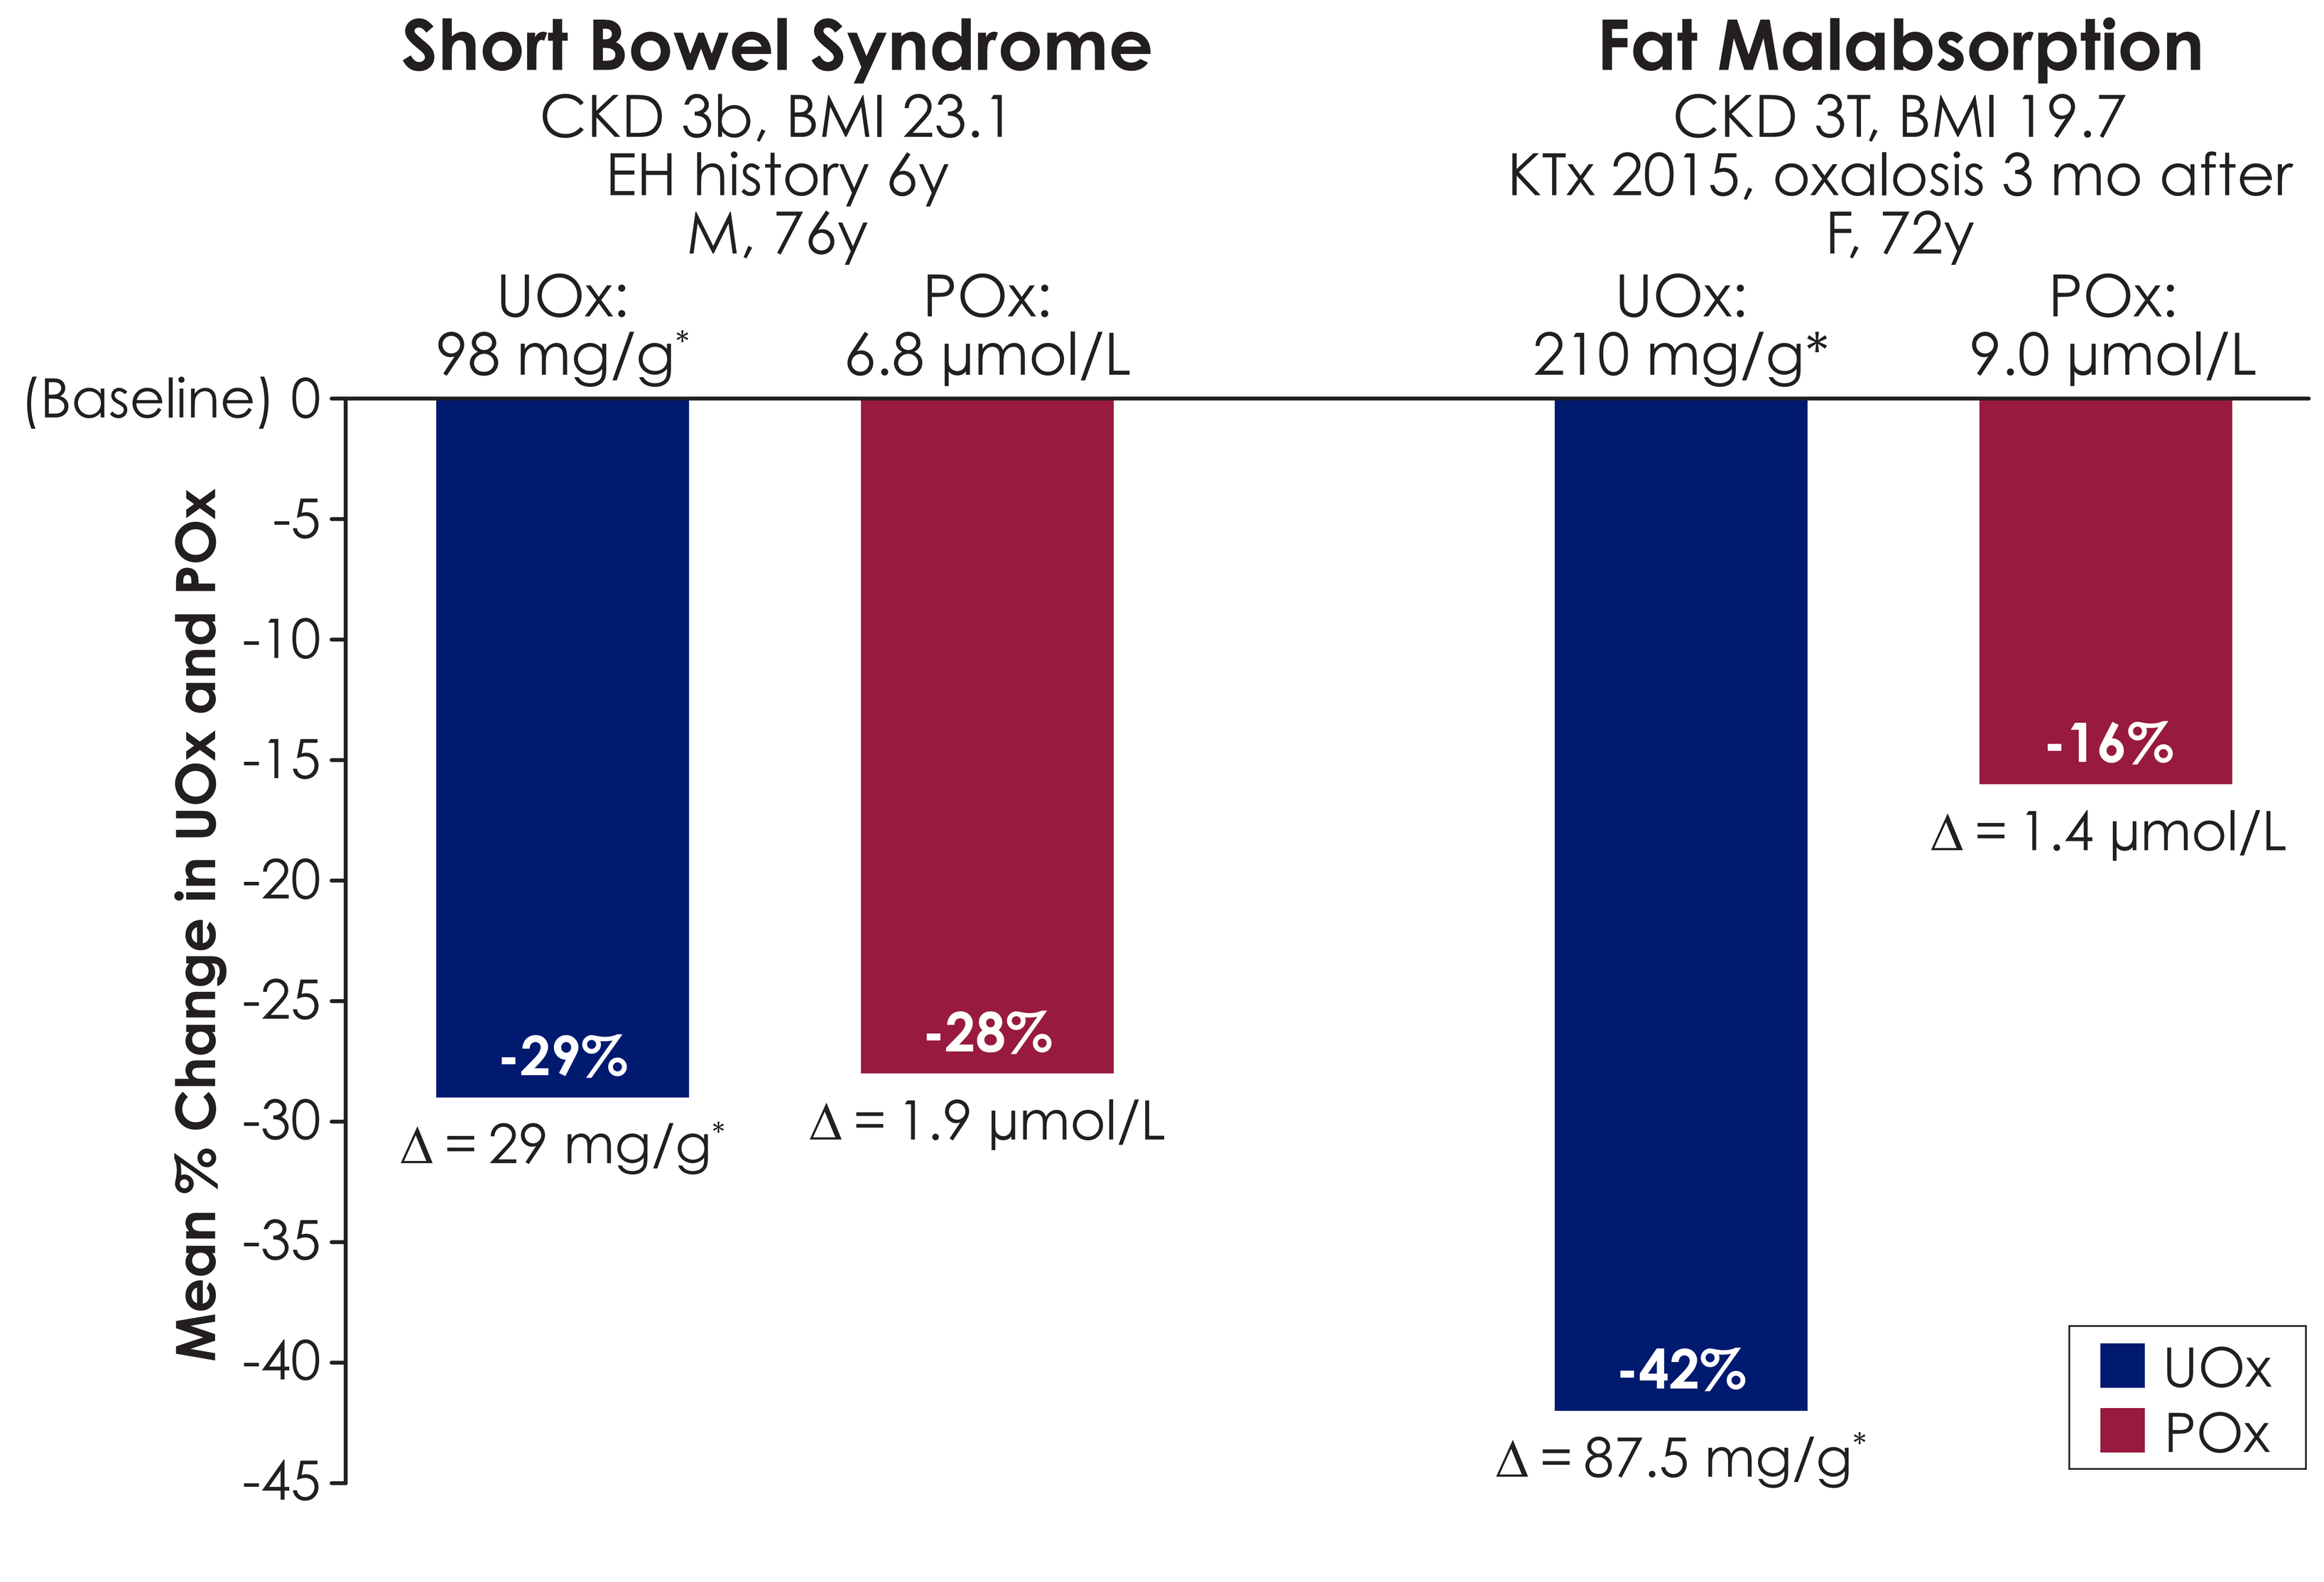
Short Bowel Syndrome
CKD 3b, BMI 23.1
EH history 6y
M, 76y
UOx: 98 mg/g*
POx: 6.8 µmol/L
Fat Malabsorption
CKD 3T, BMI 19.7
KTx 2015, oxalosis 3 mo after
F, 72y
UOx: 210 mg/g*
POx: 9.0 µmol/L
(Baseline) 0
-5
-10
-15
-20
-25
-30
-35
-40
-45
Mean % Change in UOx and POx
-29%
Δ = 29 mg/g*
-28%
Δ = 1.9 µmol/L
-42%
Δ = 87.5 mg/g*
-16%
Δ = 1.4 µmol/L
UOx
POx

*Urinary oxalate (UOx mg/d) was normalized to creatine

Plasma Oxalate Reduction in Subjects on Dialysis

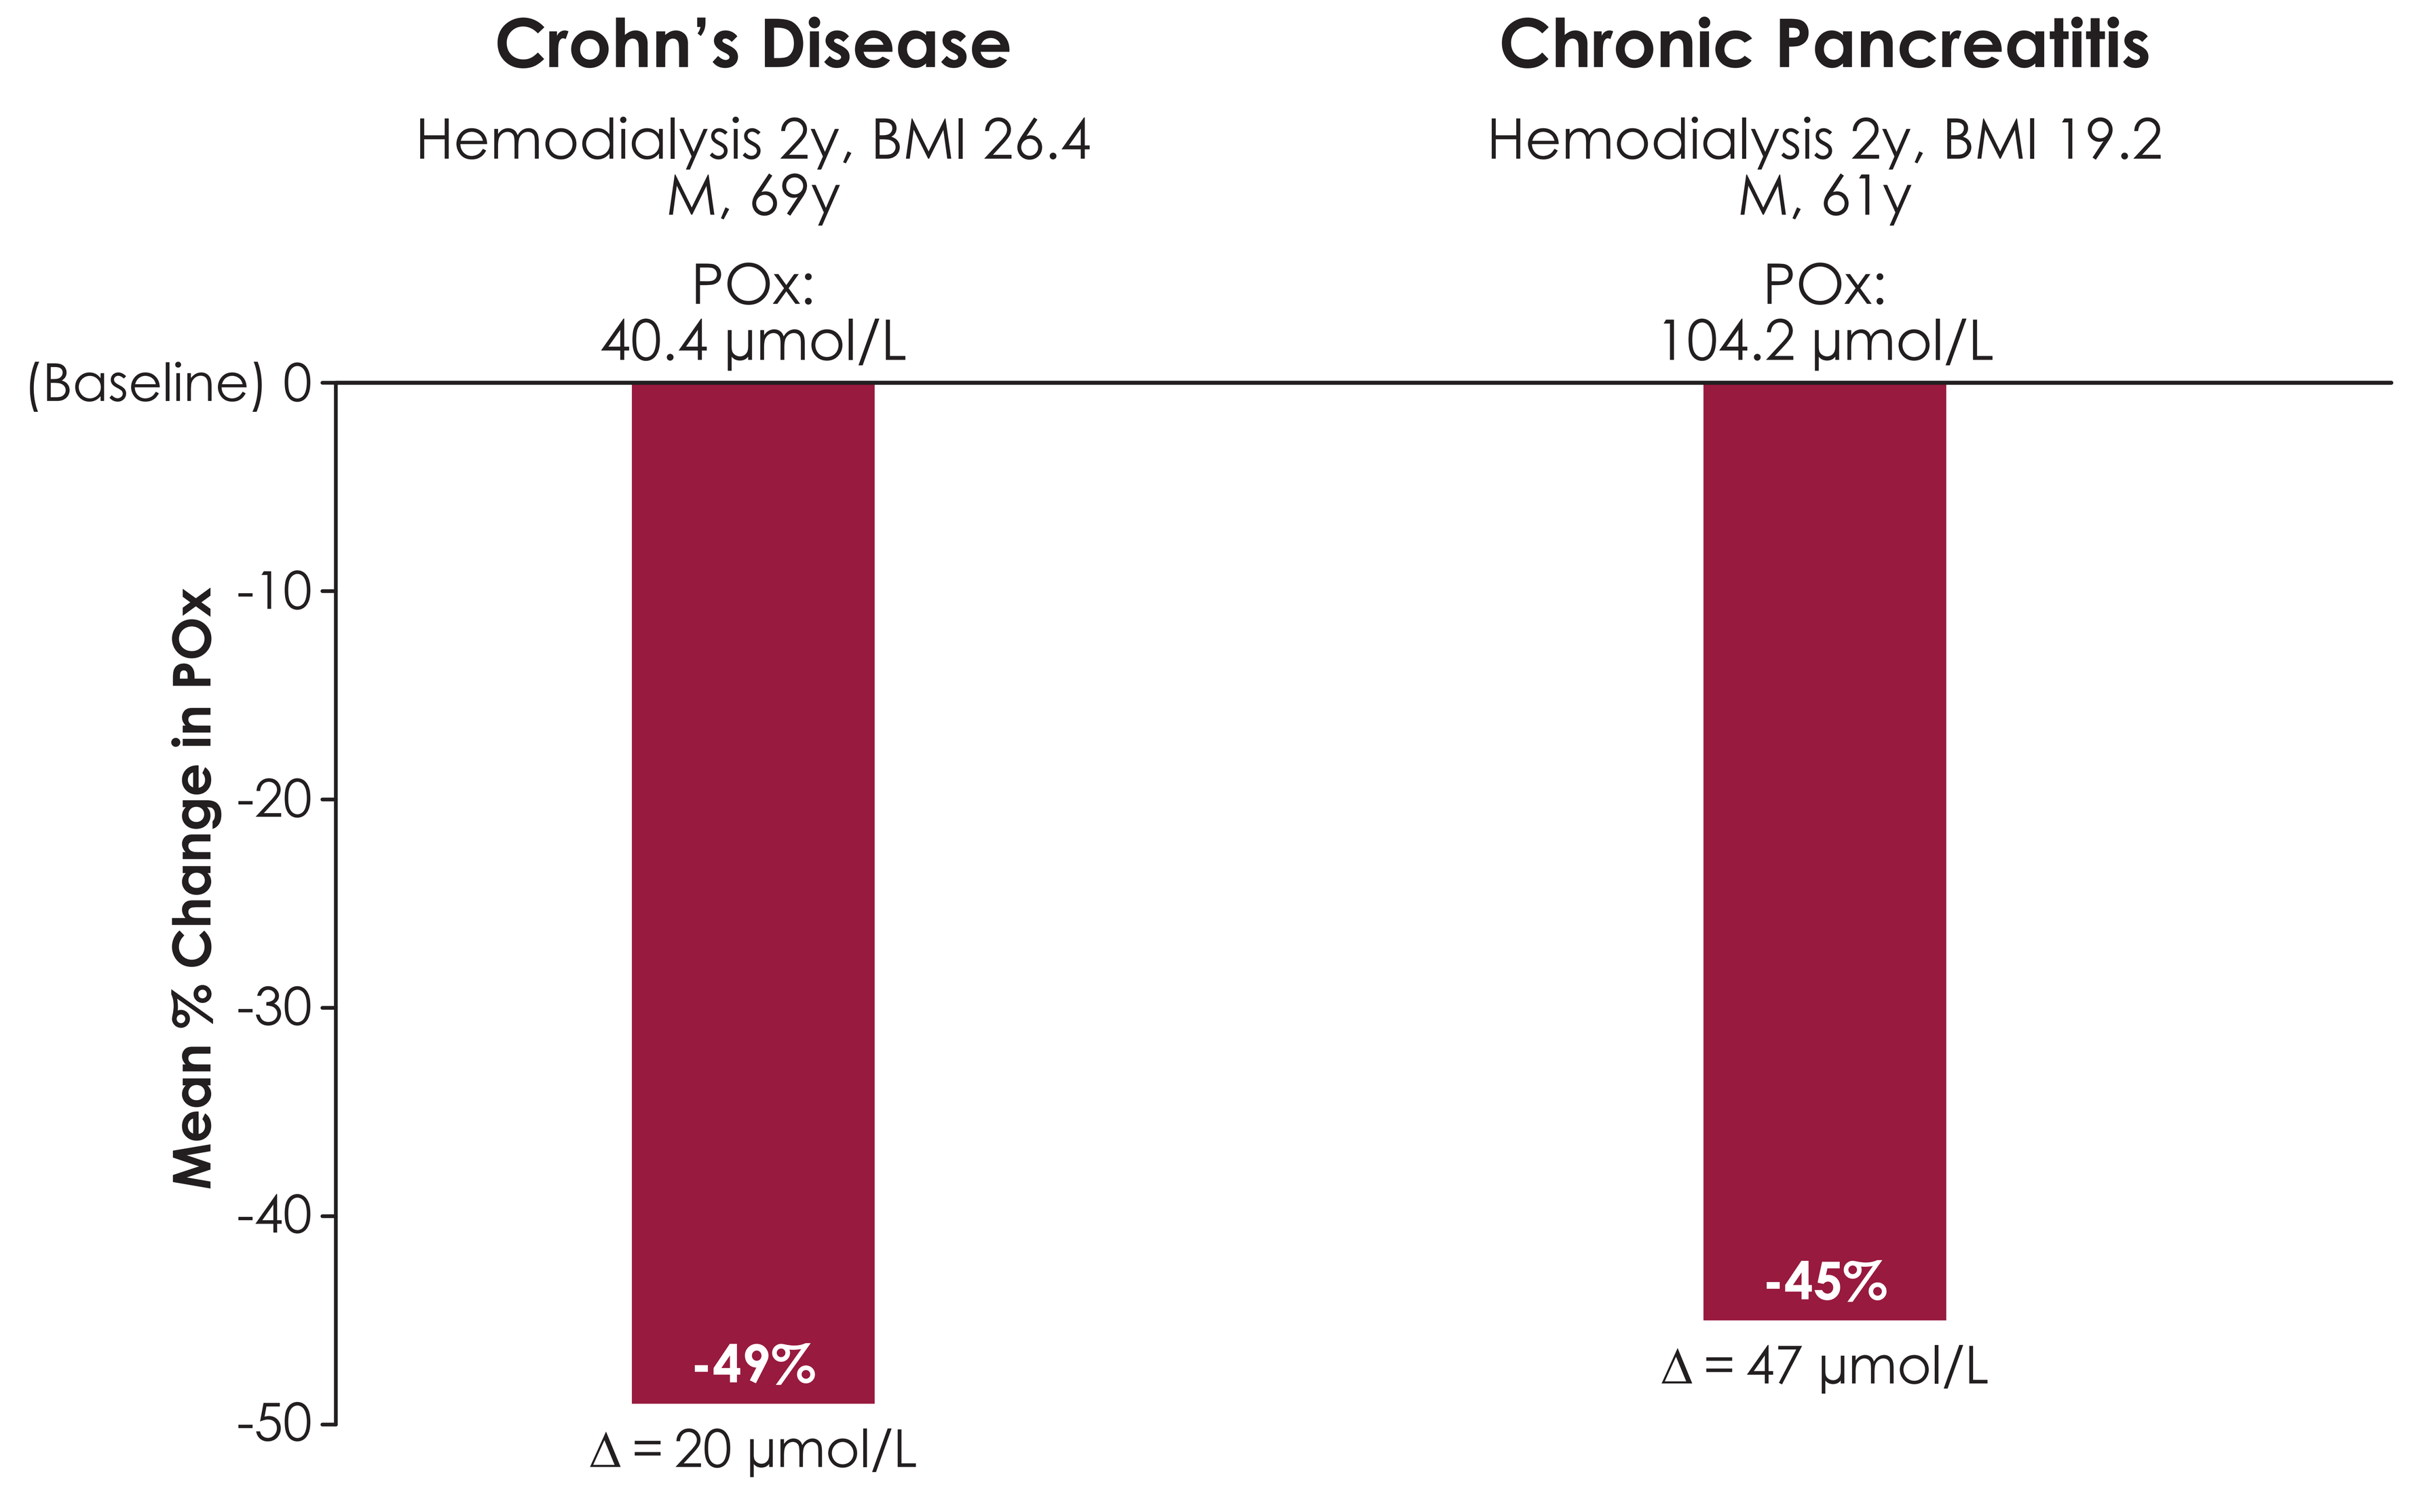
Crohn's Disease
Hemodialysis 2y, BMI 26.4
M, 69y
POx: 40.4 µmol/L
Chronic Pancreatitis
Hemodialysis 2y, BMI 19.2
M, 61y
POx: 104.2 µmol/L
(Baseline) 0
-10
-20
-30
-40
-50
Mean % Change in POx
-49%
Δ = 20 µmol/L
-45%
Δ = 47 µmol/L

POx upper limit of normal: < 2 µmol/L Supersaturation threshold: 30 µmol/L

Summary

- Reloxaliase was well tolerated and reduced both UOx and POx, suggesting the potential for reducing systemic oxalate deposition with chronic therapy
- Reduction in plasma oxalate, if sustained, can potentially delay need for dialysis, reduce the frequency of dialysis sessions, help qualify for kidney transplant or protect a new kidney post-transplant
- These initial data support further testing of reloxaliase in patients with severe EH and CKD with hyperoxalemia

Acknowledgment: This research was sponsored by Allena Pharmaceuticals, and was made possible by the valuable contributions of participating investigators and patients. We want to thank all patients and families for participating, this study is ongoing and please contact us if you are interested in taking part in the study.

References: 1) Lumlertgul et al KI 2018 2) Woodcock et al NJM 2017. This poster does not present data for PH.

ClinicalTrials.gov Identifier: NCT03391804

For information about recruitment and participating research centers, please call 617-467-4577 x379 or email clinical206@allenapharma.com